Local Regulated IL-12 Expression as an Immunotherapy for the Treatment of Pontine Glioma

John A. Barrett[1], Hongliang Cai[1], John Miao[1], Karah Ludington[2], Courtney Devore[2], Paul Gonzales[2], Sam Broder[3], Francois Lebel[1], Laurence JN Cooper[1]
[1]Ziopharm Oncology Inc., Boston, MA, United States, 02129
[2]Translational Drug Development, Scottsdale, AZ, United States, 85259
[3]Formerly of Intrexon Corporation, Germantown, MD, United States, 20876

BACKGROUND

- Glioma in the pontine region accounts for ~15% of pediatric brain tumors with a grim median survival time of <1 year. Immunotherapy may be an attractive approach for the treatment of pontine glioma, however, the lack of defined tumor antigens with homogeneous expression that may be safely targeted has handicapped this approach.
- We are conducting clinical trials to recruit and stimulate the endogenous immune response by local delivery of a replication-incompetent adenovirus engineered to conditionally express IL-12, via our RheoSwitch Therapeutic System® (RTS®) gene switch (Ad-RTS-IL-12).
- Previous studies in supratentorial syngeneic mouse tumor models demonstrated that Ad-RTS-mIL-12 + veledimex elicited a dose-related increase in tumor IL-12 mRNA resulting in IL-12 protein and downstream IFN-γ which correlated with increased anti-tumor effects and survival. In ongoing clinical trials with Ad-RTS-hIL-12 + veledimex within recurrent supratentorial gliomas, we observed encouraging improvement in overall survival and toxicity profile that is readily manageable by stopping veledimex.

TECHNOLOGY

Inducible Gene Regulation: RheoSwitch Therapeutic System®

RheoSwitch Therapeutic System® (RTS®) is a 3-component transcriptional regulator

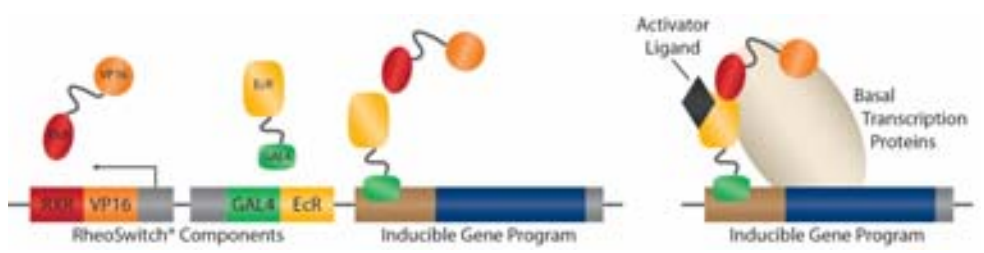
Activator Ligand
Basal Transcription Proteins
VP16
GAL4
EcR
RheoSwitch® Components
Inducible Gene Program
Inducible Gene Program

1. **The Switch Components**: The RTS® gene program includes 2 receptor protein fusions: VP16-RXR (co-activation partner, CAP) and Gal4-EcR (ligand-inducible transcription factor, LTF). In the absence of ligand, the LTF binds to the inducible promoter and does not form a stable interaction with the CAP.
2. **The Inducible Promoter**: A customizable promoter to which basal transcription proteins are recruited and the target gene is transcribed.
3. **The Activator Ligand (veledimex):** An ecdysone analog, diacylhydrazine-based small molecule functions as an activator. In the presence of the ligand, a conformational change in the LTF leads to a stable, high affinity interaction with the CAP.

PROTOCOL

GL261 Pontine Model

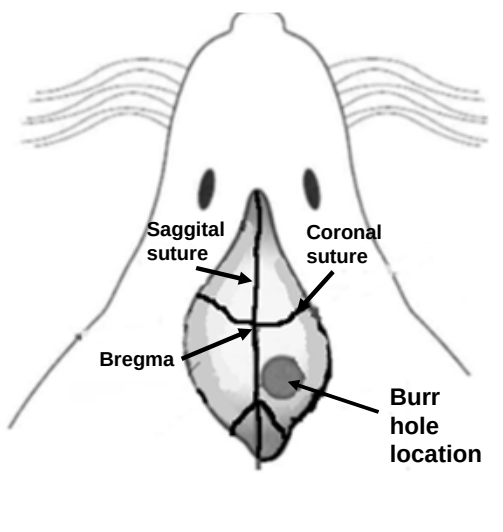
Saggital suture
Coronal suture
Bregma
Burr hole location

- Five Days prior to therapy 1 x 10^5 GL-261 glioma cells volume 3ul were administered into the pons region of the brain of C57BL/6 mice.
- On Day 1 a single dose of Ad-RTS-mIL-12 at 5x10^9vp 5 µl 1.5mm (5mm caudal Bregma) and 1mm lateral, depth of 3.5mm (PONS) followed by the activator ligand, veledimex administered orally for 14 consecutive days.
- Control therapies were administered at the dose and schedule depicted.
- The time to disease progression and death was studied.
- Tumor and serum cytokines were evaluated with ELISA.
- All values expressed as mean ± SEM
- Ad = Ad-RTS-mIL-12; V = veledimex

RESULTS

Veledimex crosses BBB in GL-261 Mouse Glioma Model

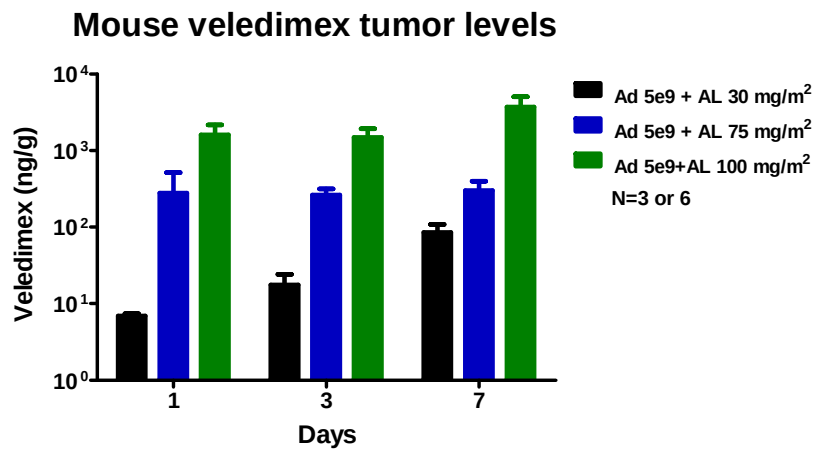
Mouse veledimex tumor levels
Veledimex (ng/g)
10⁰
10¹
10²
10³
10⁴
1
3
7
Days
Ad 5e9 + AL 30 mg/m²
Ad 5e9 + AL 75 mg/m²
Ad 5e9+AL 100 mg/m²
N=3 or 6

Veledimex dose-response: tumor and serum cytokines in GL261 Pontine Model

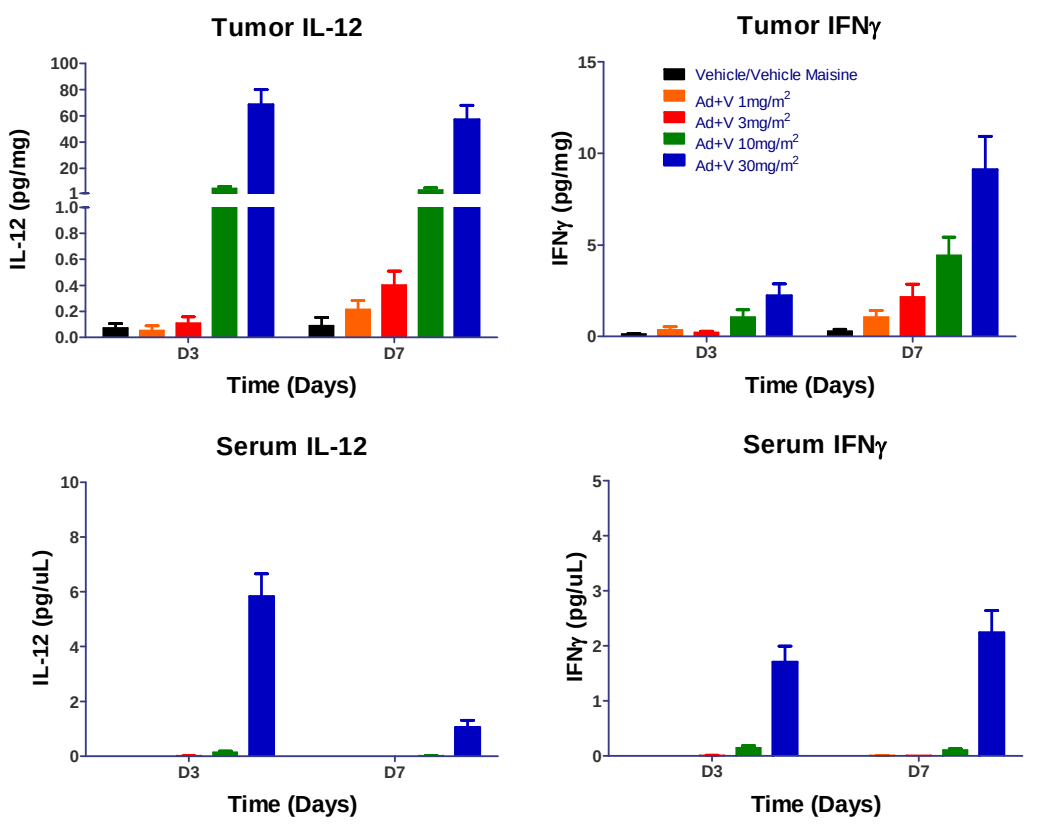
Tumor IL-12
IL-12 (pg/mg)
Time (Days)
D3
D7
Tumor IFNγ
IFNγ (pg/mg)
Vehicle/Vehicle Maisine
Ad+V 1mg/m²
Ad+V 3mg/m²
Ad+V 10mg/m²
Ad+V 30mg/m²
Serum IL-12
IL-12 (pg/uL)
Serum IFNγ
IFNγ (pg/uL)

Ad-RTS-mIL-12 + Veledimex Superior Efficacy In Pontine Mouse Model

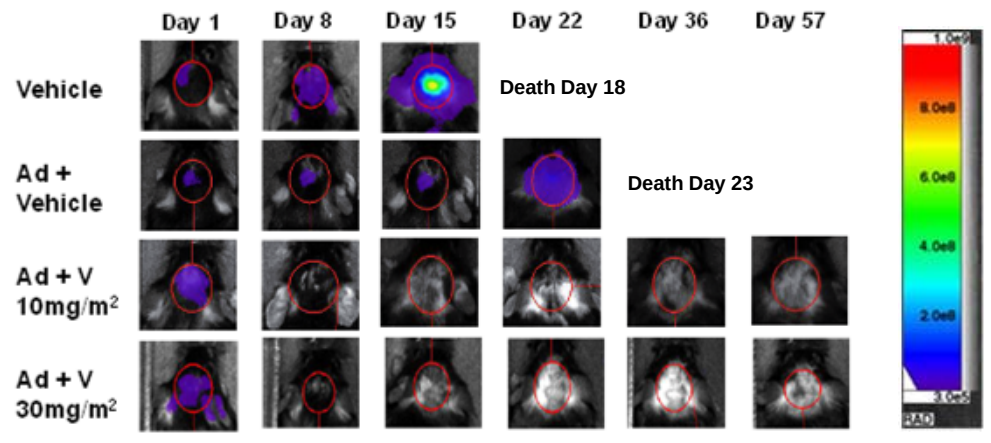
Day 1
Day 8
Day 15
Day 22
Day 36
Day 57
Vehicle
Death Day 18
Ad + Vehicle
Death Day 23
Ad + V 10mg/m²
Ad + V 30mg/m²

1e5 GL261 red-luc cells, 5 days later, begin therapy

RESULTS

Ad-RTS-mIL-12 + Veledimex Results in Increased Survival in Orthotopic Pontine Glioma Mouse Model

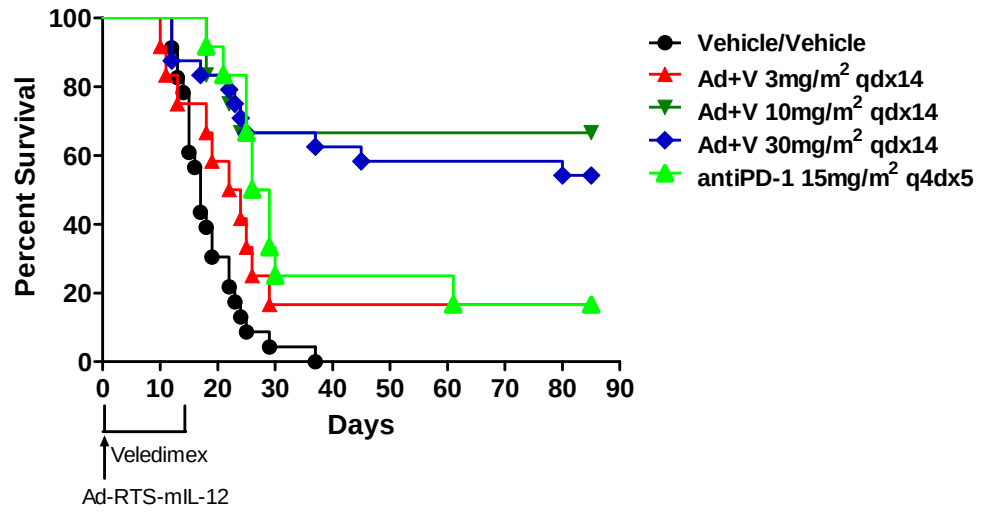
Percent Survival
Days
Vehicle/Vehicle
Ad+V 3mg/m² qdx14
Ad+V 10mg/m² qdx14
Ad+V 30mg/m² qdx14
antiPD-1 15mg/m² q4dx5
Veledimex
Ad-RTS-mIL-12

Ad-RTS-mIL-12 + Veledimex Results in Tumor Free Long Term Survival

Treatment	Dose	Number of Mice with Tumors at Moribund Sacrifice (Tumor/Total Mice)	Number of Long Term Survivors with No Tumor (No Tumor/Total Survivors)
PBS Vehicle + Maisine Vehicle	0 mg/kg + 0 mg/kg	22/22	0/0
Ad-RTS-mIL-12 + Veledimex	5x10^9 vp + 3 mg/m^2	10/12	2/2
Ad-RTS-mIL-12 + Veledimex	5x10^9 vp + 10 mg/m^2	4/12	8/8
Ad-RTS-mIL-12 + Veledimex	5x10^9 vp + 30 mg/m^2	6/22	13/13
antiPD-1	15 mg/m^2	10/12	2/2

Ad-RTS-mIL-12 + Veledimex Change in Body Weight

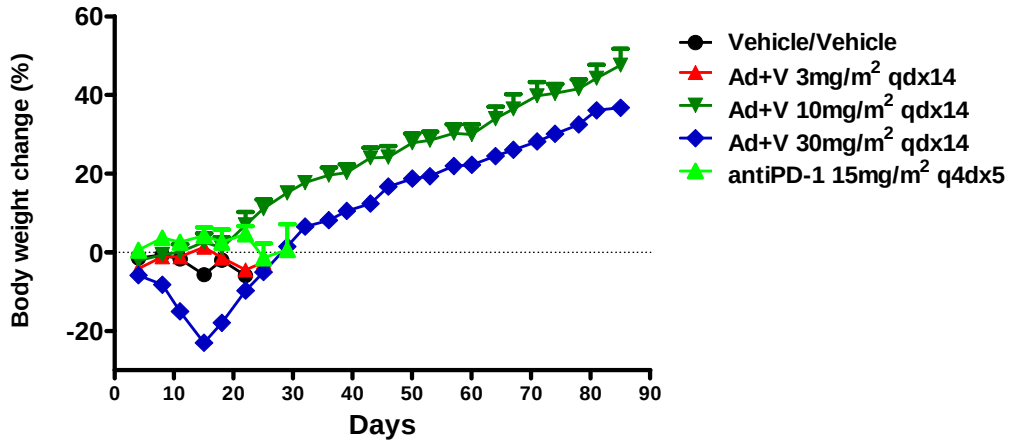
Body weight change (%)
Days
Vehicle/Vehicle
Ad+V 3mg/m² qdx14
Ad+V 10mg/m² qdx14
Ad+V 30mg/m² qdx14
antiPD-1 15mg/m² q4dx5

CONCLUSIONS

- Oral veledimex exhibited dose-related increases in brain tumor exposure.
- The dose-related increase in tumor veledimex + intracranial Ad-RTS-mIL-12 resulted in a dose-related increase in tumor IL-12 and downstream IFN-γ with minimal increase in serum levels.
- In a model of pontine glioma, the controlled local tumor IL-12 production stimulated the immune system in the presence of innate tumor immunosuppression and proved to be beneficial with a profound increase in survival.
- This murine model suggests that Ad-RTS-IL-12 + veledimex 10 mg/m^2 is the optimal dose to maximize survival and therapeutic index in pontine glioma.
- **We are planning a Phase I pediatric brain tumor study design to assess Ad-RTS-IL-12 + veledimex.**

ZIOPHARM Oncology, Inc.

John A Barrett, Ph.D.
jabarrett@ziopharm.com

INTREXON®